4th BIOPHARM SCIENTIFIC ANNUAL MEETING
BSAM4, Alger le 06 juin 2024

CLINICAL TRIAL: TREATMENT OF LUNG AND LIVER CANCER WITH 188RE-IMDENDRIM

Weiyang JIN3,5, Jinde CHEN2, Hafid BELHADJ-TAHAR1,4,6, Nouredine SADEG1,4,Yong GAO2*,Guanghua YANG1,3,5,7*

1.NanoGun Technology NGT, 60230,Chambly, France
2.Department of Oncology, Shanghai East Hospital, Tongji University School of Medicine, Shanghai, PR. China.
3.International Research Center for Biological Sciences, Ministry of Science and Technology, Shanghai Ocean University, Shanghai, 201306, China.
4.Recherche & Expertise Toxicologiques (AFPreMed), 31100 Toulouse, France.
5. Zhejiang Huayi Nano Biotechnology Co., Ltd. Hangzhou, China.
6.COLCOM, Cap Alpha, Avenue de l'Europe - Clapiers, 34940 Montpellier, France
7.Zhejiang Huayi Health Industry Development Co., Ltd, Hangzhou, China. * E-mail: ghyang119@163.com

Résumé : ImDendrim treatment in solid tumor patients is highly targeted, good safety and efficacy. Indicating it hold great potential as liquid brachytherapy device candidate for solid tumors therapy.

Mots-clés: ImDendrim， solid Tumor， 188Re， CT

I- Introduction :

There are many therapeutic modalities for the tumoral treatment, but there is still no particularly effective treatment for non-operable solid tumors, particularly those of the lung and liver. In the recent past, the application of radionuclide therapy in the treatment of solid tumors has garnered increasing interest and has emerged as a novel therapeutic approach for cancer. This study aims to assess the safety, distribution, and therapeutic efficacy of ImDendrim in the management of solid tumors.

II- Matériels et méthode:

ImDendrim was administered to 36 selected patients with liver cancer (n=16) and lung cancer (n=20) by percutaneous intratumoral injection with doses 10cm2 and 20cm2. The distribution of ImDendrim and its therapeutic effect were studied using SPECT/CT. The safety of ImDendrim was retrospectively evaluated.

III- Résultats et discussion:

Our study demonstrated that ImDendrim treatment in solid tumor patients is highly targeted, good safety and efficacyThe nitroimidazole in ImDendrim is designed to target hypoxic tumour cells, while the β-rays emitted by 188Re are employed to kill tumours. Furthermore, the γ-rays of 188Re facilitate the integration of diagnosis and treatment.

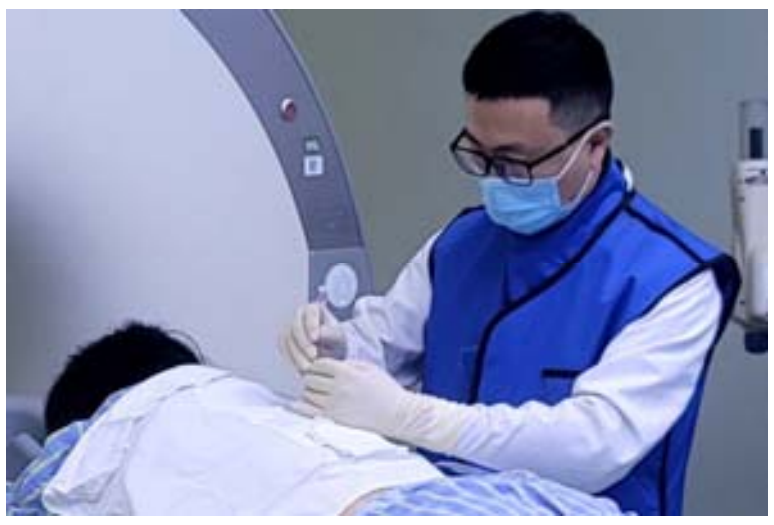

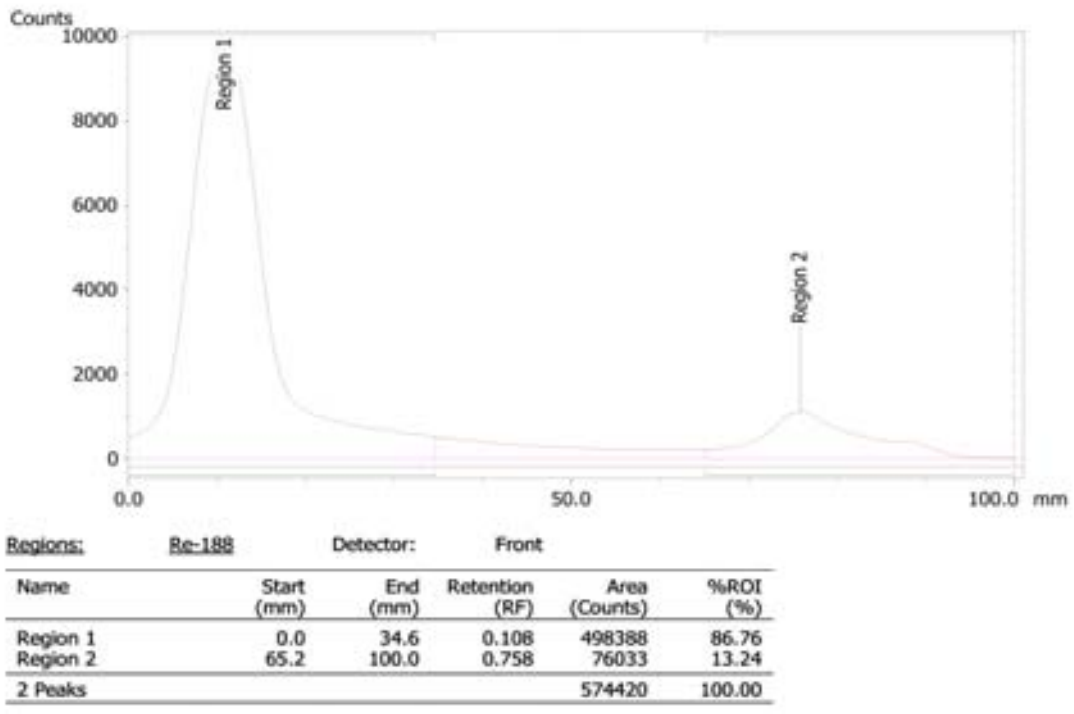
Counts
10000
8000
6000
4000
2000
0
0.0
50.0
100.0 mm
Region 1
Region 2
Regions: Re-188 Detector: Front
Name Start (mm) End (mm) Retention (RF) Area (Counts) %ROI (%)
Region 1 0.0 34.6 0.108 498388 86.76
Region 2 65.2 100.0 0.758 76033 13.24
2 Peaks 574420 100.00

Figure 1: ImDendrim Radiochemical Purity

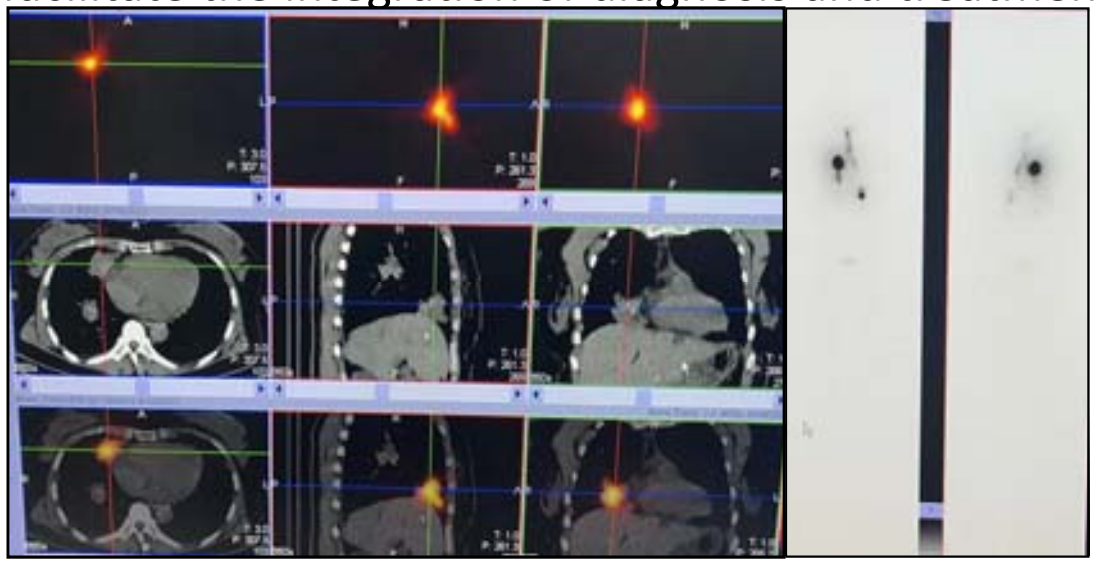

Figure 2: The biodistribution of imDendrim was imaged using SPECT with γ rays emitted by 188Re.

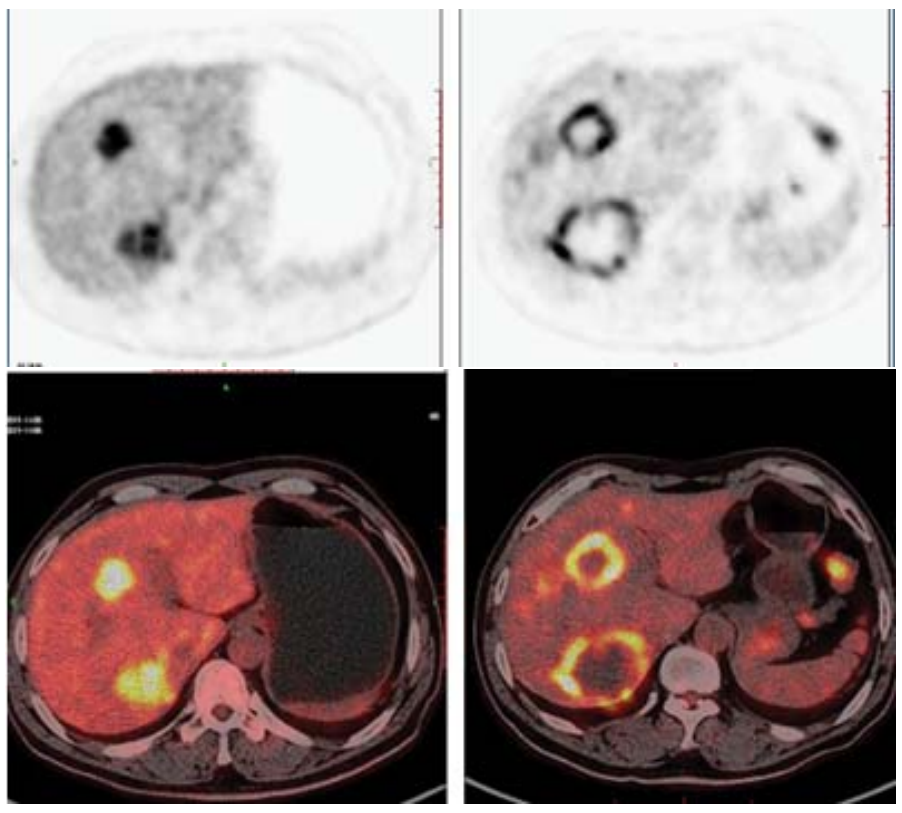

Before treatment After treatment

Urine within 24 hours						Total
urine volume	0(base line)	1mL	2 mL	5 mL	10 mL	2210 mL
Activity (μ Ci)	210	219	227	251	287	17680
count	173	1141	1981	4333	6900	$1.68X10^{6}$

Urine within 48 hours						Total
urine volume	0(base line)	1mL	2 mL	5 mL	10 mL	2210 mL
Activity (μ Ci)	49	49	49	50	52	263
count	87	101	115	230	378	51876

Figure 3: Urine monitoring

Conclusion:Indicating ImDendrim hold great potential as liquid brachytherapy device candidate for solid tumors therapy.

Références： (1)Yang Guanghua, Sadeg Nouredine and Hafid Belhadj-Tahar. New Potential In Situ Anticancer Agent Derived from [188Re]rhenium Nitro-Imidazole Ligand Loaded 5th Generation Poly-L-Lysine Dendrimer for Treatment of Transplanted Human Liver Carcinoma in Nude Mice. Drug Designing 2017 , 6 (1):1-7.